In vitro antiprotozoal action of 7 spices, medicinal plants and essential oils against 3 stages of Eimeria tenella

Claire Girard, Anne Silvestre, Thibaut Chabrillat, Sylvain Kerros

▶ **To cite this version:**

Claire Girard, Anne Silvestre, Thibaut Chabrillat, Sylvain Kerros. In vitro antiprotozoal action of 7 spices, medicinal plants and essential oils against 3 stages of Eimeria tenella. World's Poultry Congress (WPC), 2021, Online, France. hal-04126107

HAL Id: hal-04126107

https://hal.inrae.fr/hal-04126107

Submitted on 13 Jun 2023

HAL is a multi-disciplinary open access archive for the deposit and dissemination of scientific research documents, whether they are published or not. The documents may come from teaching and research institutions in France or abroad, or from public or private research centers.

L'archive ouverte pluridisciplinaire **HAL**, est destinée au dépôt et à la diffusion de documents scientifiques de niveau recherche, publiés ou non, émanant des établissements d'enseignement et de recherche français ou étrangers, des laboratoires publics ou privés.

Distributed under a Creative Commons Attribution - NonCommercial 4.0 International License

In vitro antiprotozoal action of 7 spices, medicinal plants and essential oils against 3 stages of *Eimeria tenella*

C. GIRARD(1), A. SILVESTRE(2), T. CHABRILLAT(1), S. KERROS(1).
(1)PHYTOSYNTHESE, F-63200 MOZAC, FRANCE
(2)INRAE, UNIVERSITÉ DE TOURS, ISP, F-37380 NOUZILLY, FRANCE

Growing societal concerns to replace coccidiostats in poultry farms have led to the use of alternatives such as plant bio-actives. To investigate the mode of action of natural molecules, we designed this in vitro study to screen anti-protozoal activity of 4 spices or medicinal plants described by non-volatile active molecules (MedP) and 3 essential oils (EO) against 3 different stages of E. tenella.

MATERIAL AND METHODS

- **Natural products tested**:
 - **MedP**: Fabacea powder, Urticaceae extract, Punicaceae extract, Acanthaceae extract
 - **EO**: Lilaceae EO, Lamiaceae EO, Lauraceae EO
- **Negative control**: H_20, soybean hemicellulose, DMSO
- **Protozoal strain**: *Eimeria tenella* recombined strain
- 3 experimental steps after cytotoxicity assay (colorimetric MTS test, $1{,}5.10^4$ MDBK seed, incubation 5h at 37°C):
 1. Direct anti-protozoite activity test: incubation 41°C for 1h. Killed sporozoites were stained with Evans Blue Dye. Each sample tested in triplicate.
 Viability rate with fluorescence microscopy as % viability = number of live parasite in tested sample/ number of live parasite in negative control*100
 2. Invasion inhibition test: 96 microplates: 1.5×10^4 MDBK cells, 1.5×10^4 *E. tenella* sporozoites expressed enzymatic activity; substrate added for 2h at 37°C, 4 replicates per tested sample.
 % invasion = absorbance (550 nm) tested sample/ absorbance (550 nm) negative control*100
 3. Sporulation rate test: 5.10^5 non-sporulated oocysts (in 2% potassium dichromate solution), incubation for 72h at 26°C; each sample tested in triplicate.
 % sporulation = number of sporulated oocysts in tested sample/number of sporulated oocyst in negative control*100
- **Statistical test**: Mann-whitney test compared to negative control for each test ($p<0.05$).

RESULTS AND DISCUSSION

The 4 MedP samples (Fabacea powder, Urticaceae extract, Punicaceae extract, Acanthaceae extract) were less cytotoxic (lethal dose 10% (LD10) than the 3 EO samples (Lilaceae EO, Lamiaceae EO, Lauraceae EO) (MedP diluted at 10^{-2}, EO diluted at 10^{-4}-10^{-6}).

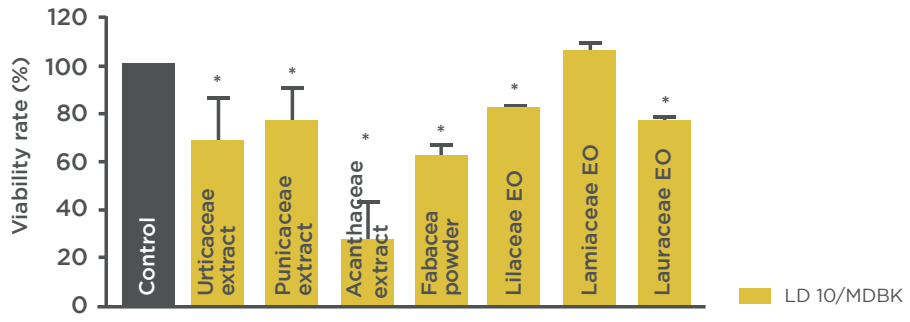
Viability rate (%)
120
100
80
60
40
20
0
Control
Urticaceae extract
Punicaceae extract
Acanthaceae extract
Fabacea powder
Lilaceae EO
Lamiaceae EO
Lauraceae EO
LD 10/MDBK

Fig.1 DIRECT ANTI-PROTOZOAL ACTIVITY AGAINST *E. TENELLA* SPOROZOITES (*: p<0.05 ; **:p<0.01; *: p<0.001)**

MedP samples showed direct anti-protozoal activity against *E. tenella* stages: sporozoite viability rates were between 76.2% and 27% (p=0.05). Lilaceae EO and Lauraceae EO exhibited 81% and 77% (p=0.05) sporozoite viability rates, respectively (Fig. 1).

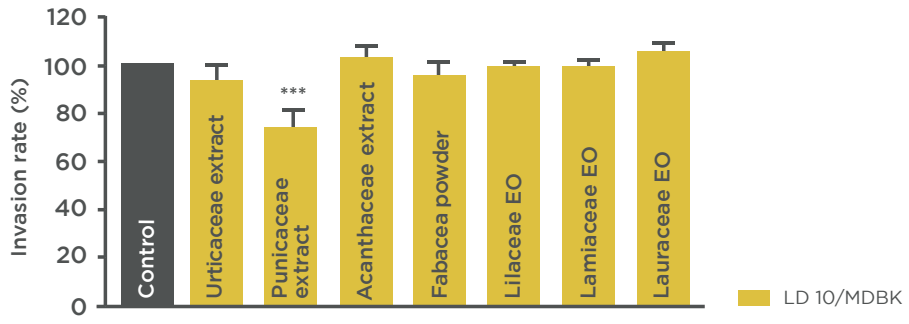
Invasion rate (%)
120
100
80
60
40
20
0

Control
Urticaceae extract
Punicaceae extract
Acanthaceae extract
Fabacea powder
Lilaceae EO
Lamiaceae EO
Lauraceae EO
LD 10/MDBK

Fig.2 MDBK CELL INVASION BY *E. TENELLA* (*: p<0.05; **: p<0.01; *: p<0.001)**

Punicaceae extract showed a reduction of 32% of cell invasion.

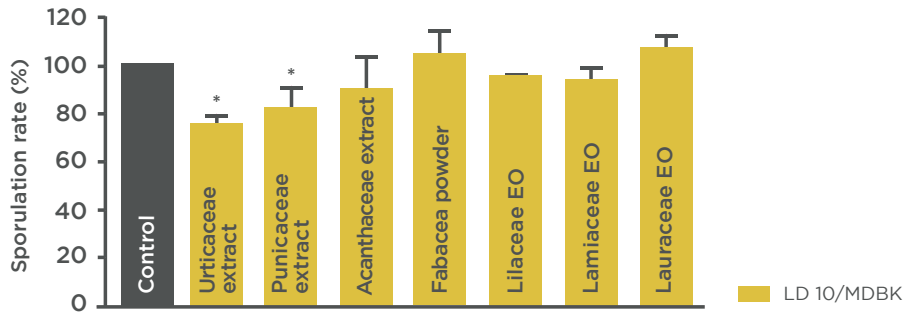
Sporulation rate (%)
120
100
80
60
40
20
0
Control
Urticaceae extract
Punicaceae extract
Acanthaceae extract
Fabacea powder
Lilaceae EO
Lamiaceae EO
Lauraceae EO
LD 10/MDBK

Fig.3 SPORULATION INHIBITION OF *E. TENELLA* (*: p<0.05; **: p<0.01; *: p<0.001)**

Acanthaceae extract and Urticaceae extract showed a sporulation inhibition of *E. tenella* oocysts (p=0.05) (Fig. 3). These medicinal plant extracts exhibited anti-protozoal activity against the different *E. tenella* stages contrary to the tested EOs which target only sporozoites. These results were in accordance with studies reporting several modes of action of medicinal plant extracts, both direct anti-protozoal activity and protective cell activity **[1]**.

CONCLUSION

Medicinal plant extracts exhibited an anti-protozoal activity against the different *E. tenella* stages. EOs only target sporozoites in this study. These findings help to understand how plant bio-actives could act to reduce *E. tenella* pathogenicity. Further studies could be carried out to test *in vitro* and/or *in vivo* potential synergies between different natural compounds.

REFERENCE

[1] WUNDERLICH et al. (2014), « Towards Identifying Novel Anti-Eimeria Agents: Trace Elements, Vitamins, and Plant-Based Natural Products ». Parasitology Research 113, no 10: 3547-56

Vegetal power & animal well-being

WWW.PHYTOSYNTHESE.COM

© Phytosynthese communication department - 2021